THE INTERACTIVE EFFECT OF PROGNOSTIC FACTORS AND PI-88 USE ON THE DELAY OF HCC RECURRENCE FOLLOWING CURATIVE PARTIAL HEPATIC RESECTION

James Garner, T Justus Homburg. Progen Pharmaceuticals, Brisbane, Australia.

PROGEN PHARMACEUTICALS

PI-88 - WELL DOCUMENTED MODE OF ACTION

- PI-88 was developed as an antagonist of heparan sulfate interaction with angiogenic growth factors (VEGF, FGF-1 and FGF-2) and as an inhibitor of heparanase that cleaves heparan sulfate (HS in the extracellular matrix (ECM), Figure. 1. Maintenance of the ECM integrity is the key to preventing tumor growth. Cleavage of HS by heparanase disrupts the ECM leading to the release of growth factors (e.g. VEGF, FGF-1 and FGF-2) which in turn promote tumor growth
- To date over 23,300 doses of the drug have been administered to over 470 patients and healthy volunteers
- Six Phase 1 and six Phase 2 trials have been completed. Two Phase 2 trials are ongoing with a Phase 3 trial due to begin in post-resection liver cancer in Q4 2007
- Evidence of clinical benefit has been observed with multiple cancers

Figure 1

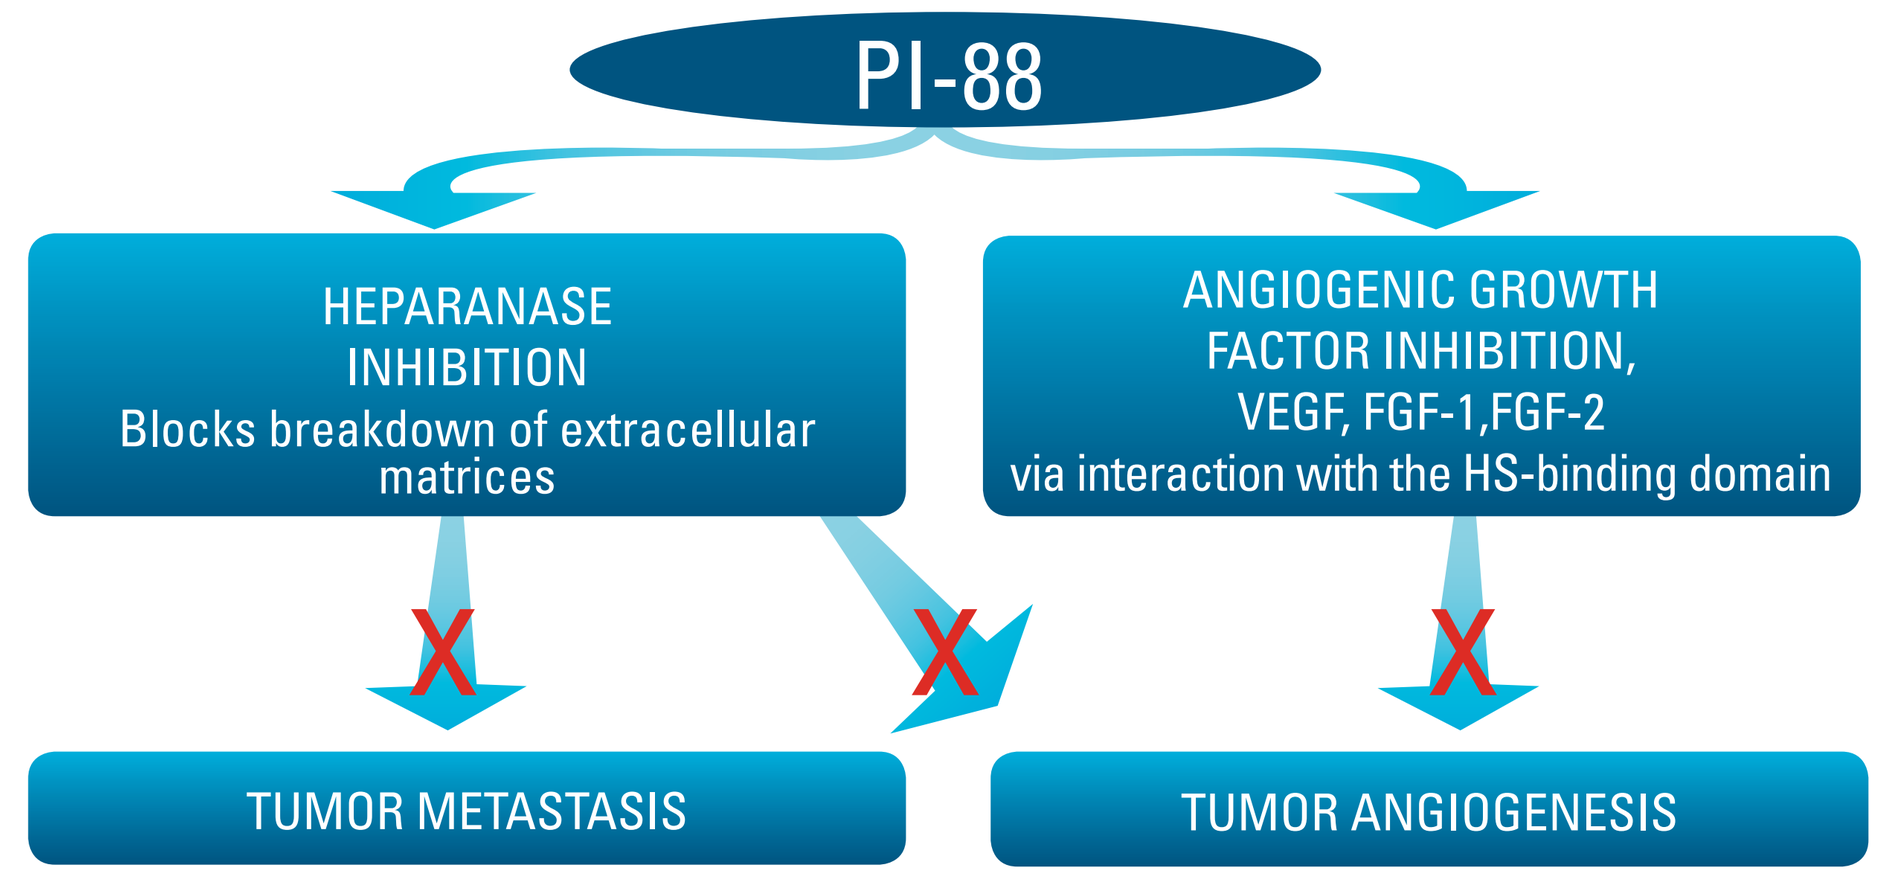
PI-88
HEPARANASE INHIBITION
Blocks breakdown of extracellular matrices
ANGIOGENIC GROWTH FACTOR INHIBITION,
VEGF, FGF-1,FGF-2
via interaction with the HS-binding domain
X
X
X
TUMOR METASTASIS
TUMOR ANGIOGENESIS

Heparan sulfate (HS) is a major component of endothelial basement membranes (BM) and extracellular matrices (ECM). PI-88 is a novel first-in-class heparan sulfate mimetic. It is believed to have anti-angiogenic and anti-metastatic effects *via* two mechanisms

CLINICAL RELEVANCE – POST-RESECTION HCC

Figure 2

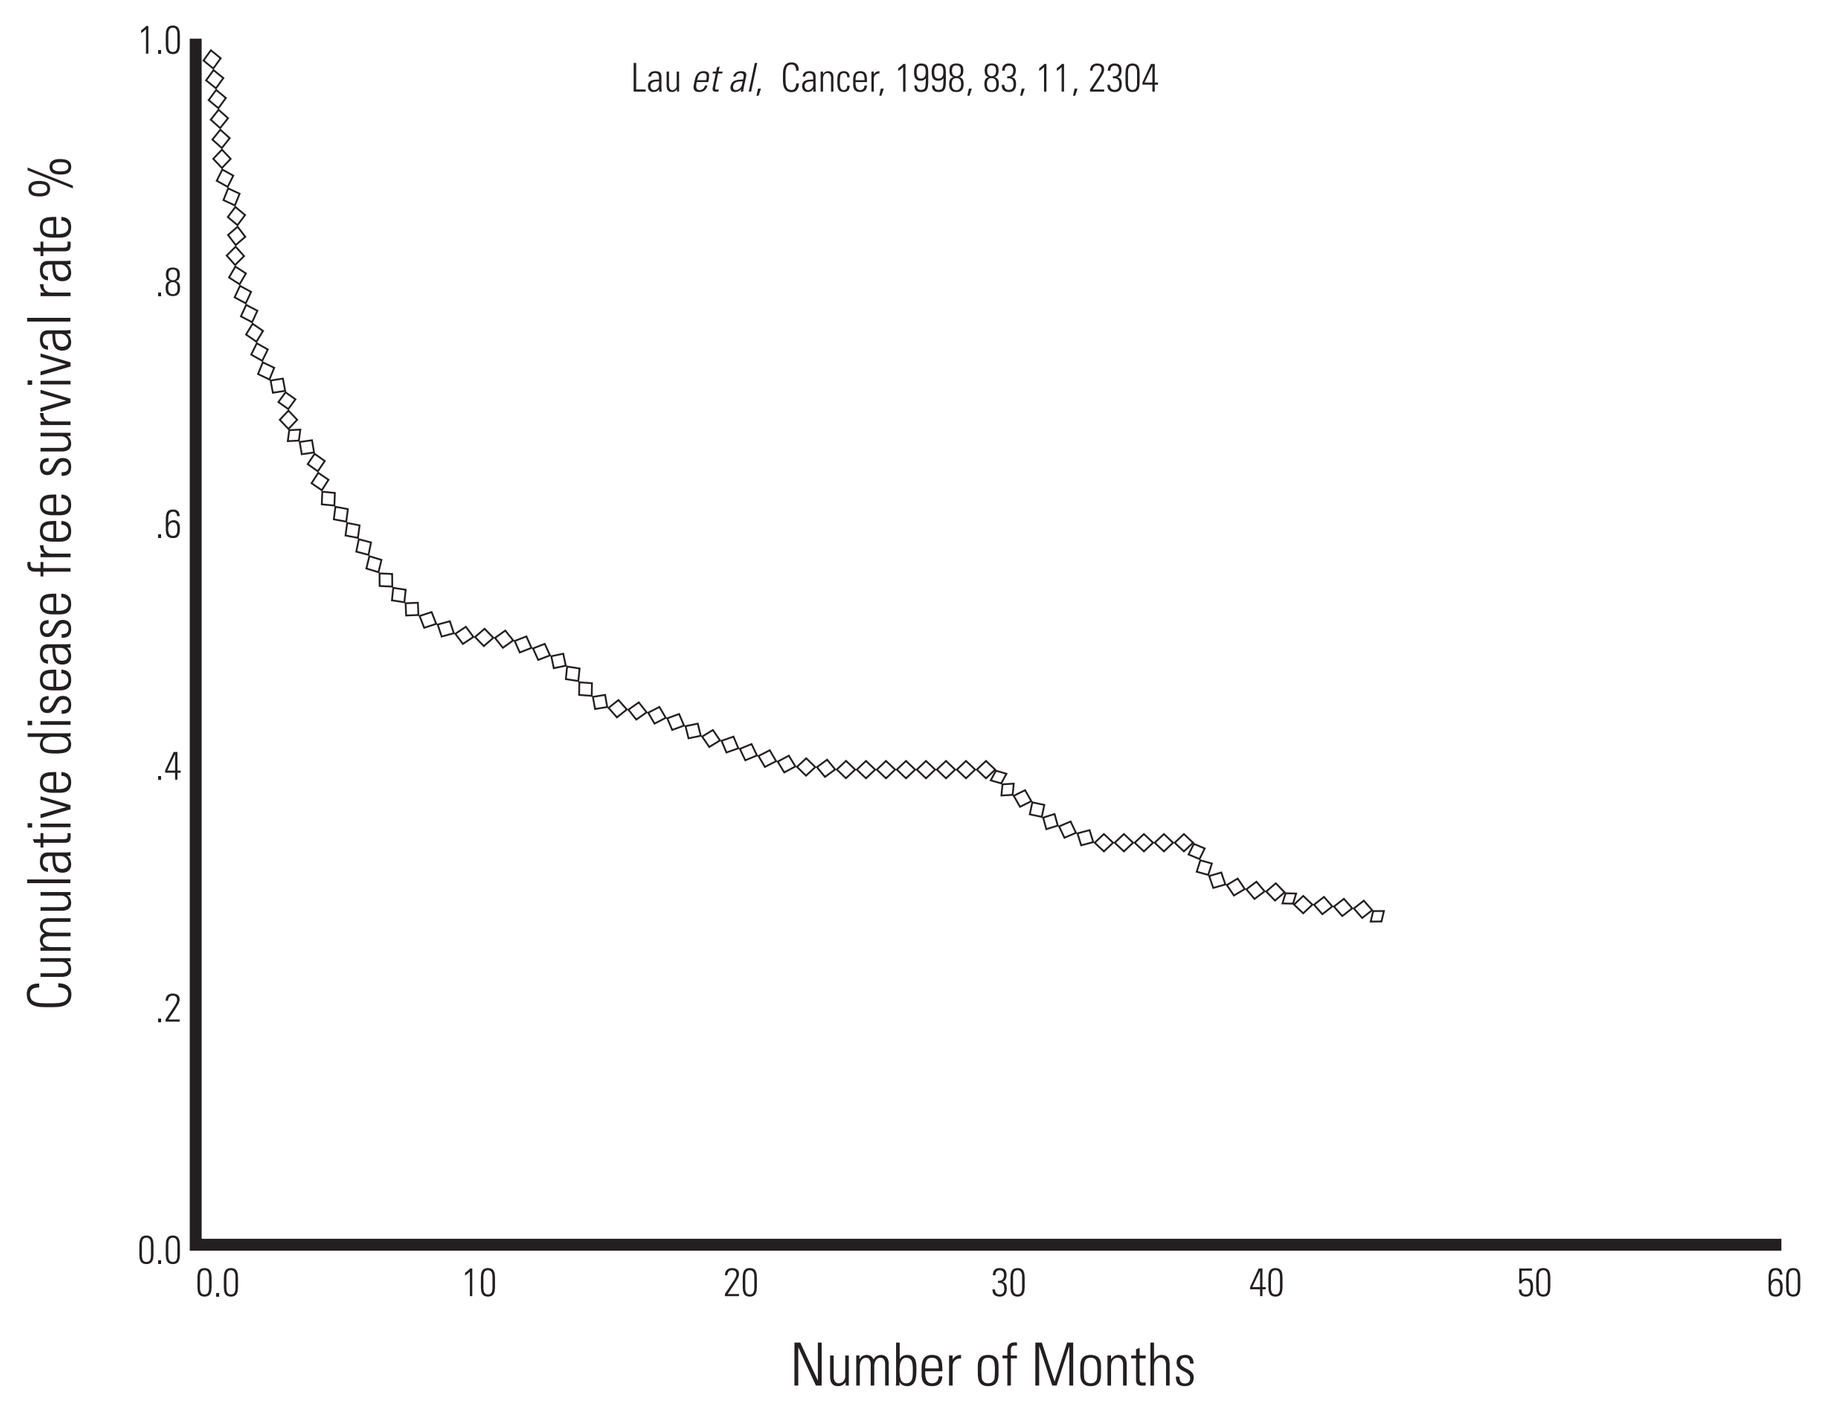
Lau et al, Cancer, 1998, 83, 11, 2304
Cumulative disease free survival rate %
1.0
.8
.6
.4
.2
0.0
0.0
10
20
30
40
50
60
Number of Months

- **Historical control reference data**
- **Median time to recurrence 12.4 months**
- **Median survival 35 months**

- Despite many studies attempting to improve treatment of post-resection liver cancer, recurrence rate remains high after resection of HCC, with 5-year cumulative recurrence rate of above 70% reported in most studies in the literature[1]

[1]Poon et al 2004, Ann. Surg. 2004;240:698-708

PURPOSE

Median time to recurrence after HCC resection is estimated at approximately 12-15 months. In the present 48-week study, approximately 50% of patients would not be expected to recur and so provide little information regarding treatment effect. This analysis examines PI-88 in "high-risk" subgroups which are more likely to recur during the study period.

METHODS

- A post hoc recursive partitioning analysis was performed to elucidate the impact of baseline prognostic variables on disease course
- Key factors for early post-operative recurrence were identified from a review of the literature
- The impact of each risk factor in isolation was tested on the control group alone
- Once the most significant factor was identified, other factors were progressively established in an iterative way
- The analysis identified a 'high-risk' population that may be expected to have undergone tumor recurrence during the 48-week duration of the study. The treatment effect of PI-88 was subsequently evaluated in this 'high-risk' population

KEY PROGNOSTIC FACTORS THAT AFFECT RECURRENCE

Table. 1

	Higher risk	Lower risk
Largest tumor size[2,3]	Larger	Smaller
Tumor number[2,4]	>1	<1
Histopathological differentiation of tumor[2,4]	Present	Absent
Microvascular invasion[2,3]	Present	Absent
Hepatitis serology[5]	HBV	HCV

[2] Poon et al. Annals of Surgery 2000; 232: 10 – 24 [3] Cha et al Journal of the American College of Surgeons 2003; 197: 753 – 758 [4] Poon et al Annals of Surgery 1999; 229: 216 – 222 [5] Sasaki et al Annals of Surgery 2006; 244(5):771-80

TRIAL DESIGN

HCC CLINICAL TRIAL PROTOCOL

Figure 3

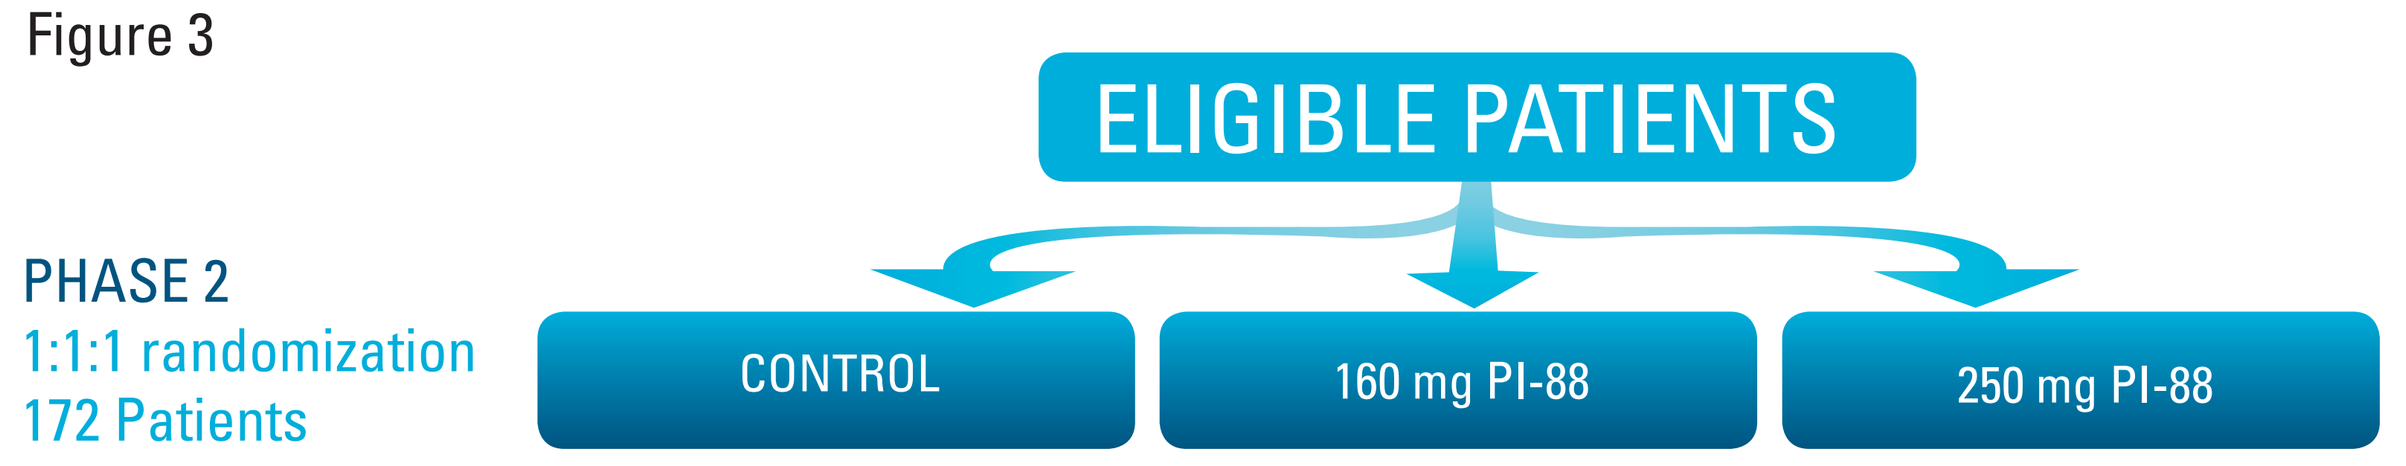
ELIGIBLE PATIENTS
PHASE 2
1:1:1 randomization
172 Patients
CONTROL
160 mg PI-88
250 mg PI-88

PRIMARY EFFICACY ENDPOINT
- Tumor non-recurrence rate within 48 weeks

SECONDARY EFFICACY ENDPOINT
- Time to first recurrence and survival rate

3-arm design comparing the control group to two dose levels of PI-88 (160 mg and 250 mg). PI-88 administered by SC injection 4 consecutive days per week doe the first 3 weeks in each 4 week cycle for up to 9 cycles (36 weeks) with a 12 week follow up period.

The drug was generally well-tolerated with injection site bruising, thrombocytopenia and asymptomatic disordered LFTs among the most common AEs, consistent with prior experience of the drug

PHASE 2 TRIAL IN POST RESECTION LIVER CANCER TOTAL SAMPLE RESULTS

In the recently completed Phase 2 randomized controlled trial, PI-88 showed evidence of efficacy at a dose of 160 mg in the post-resection adjuvant setting

Figure 4

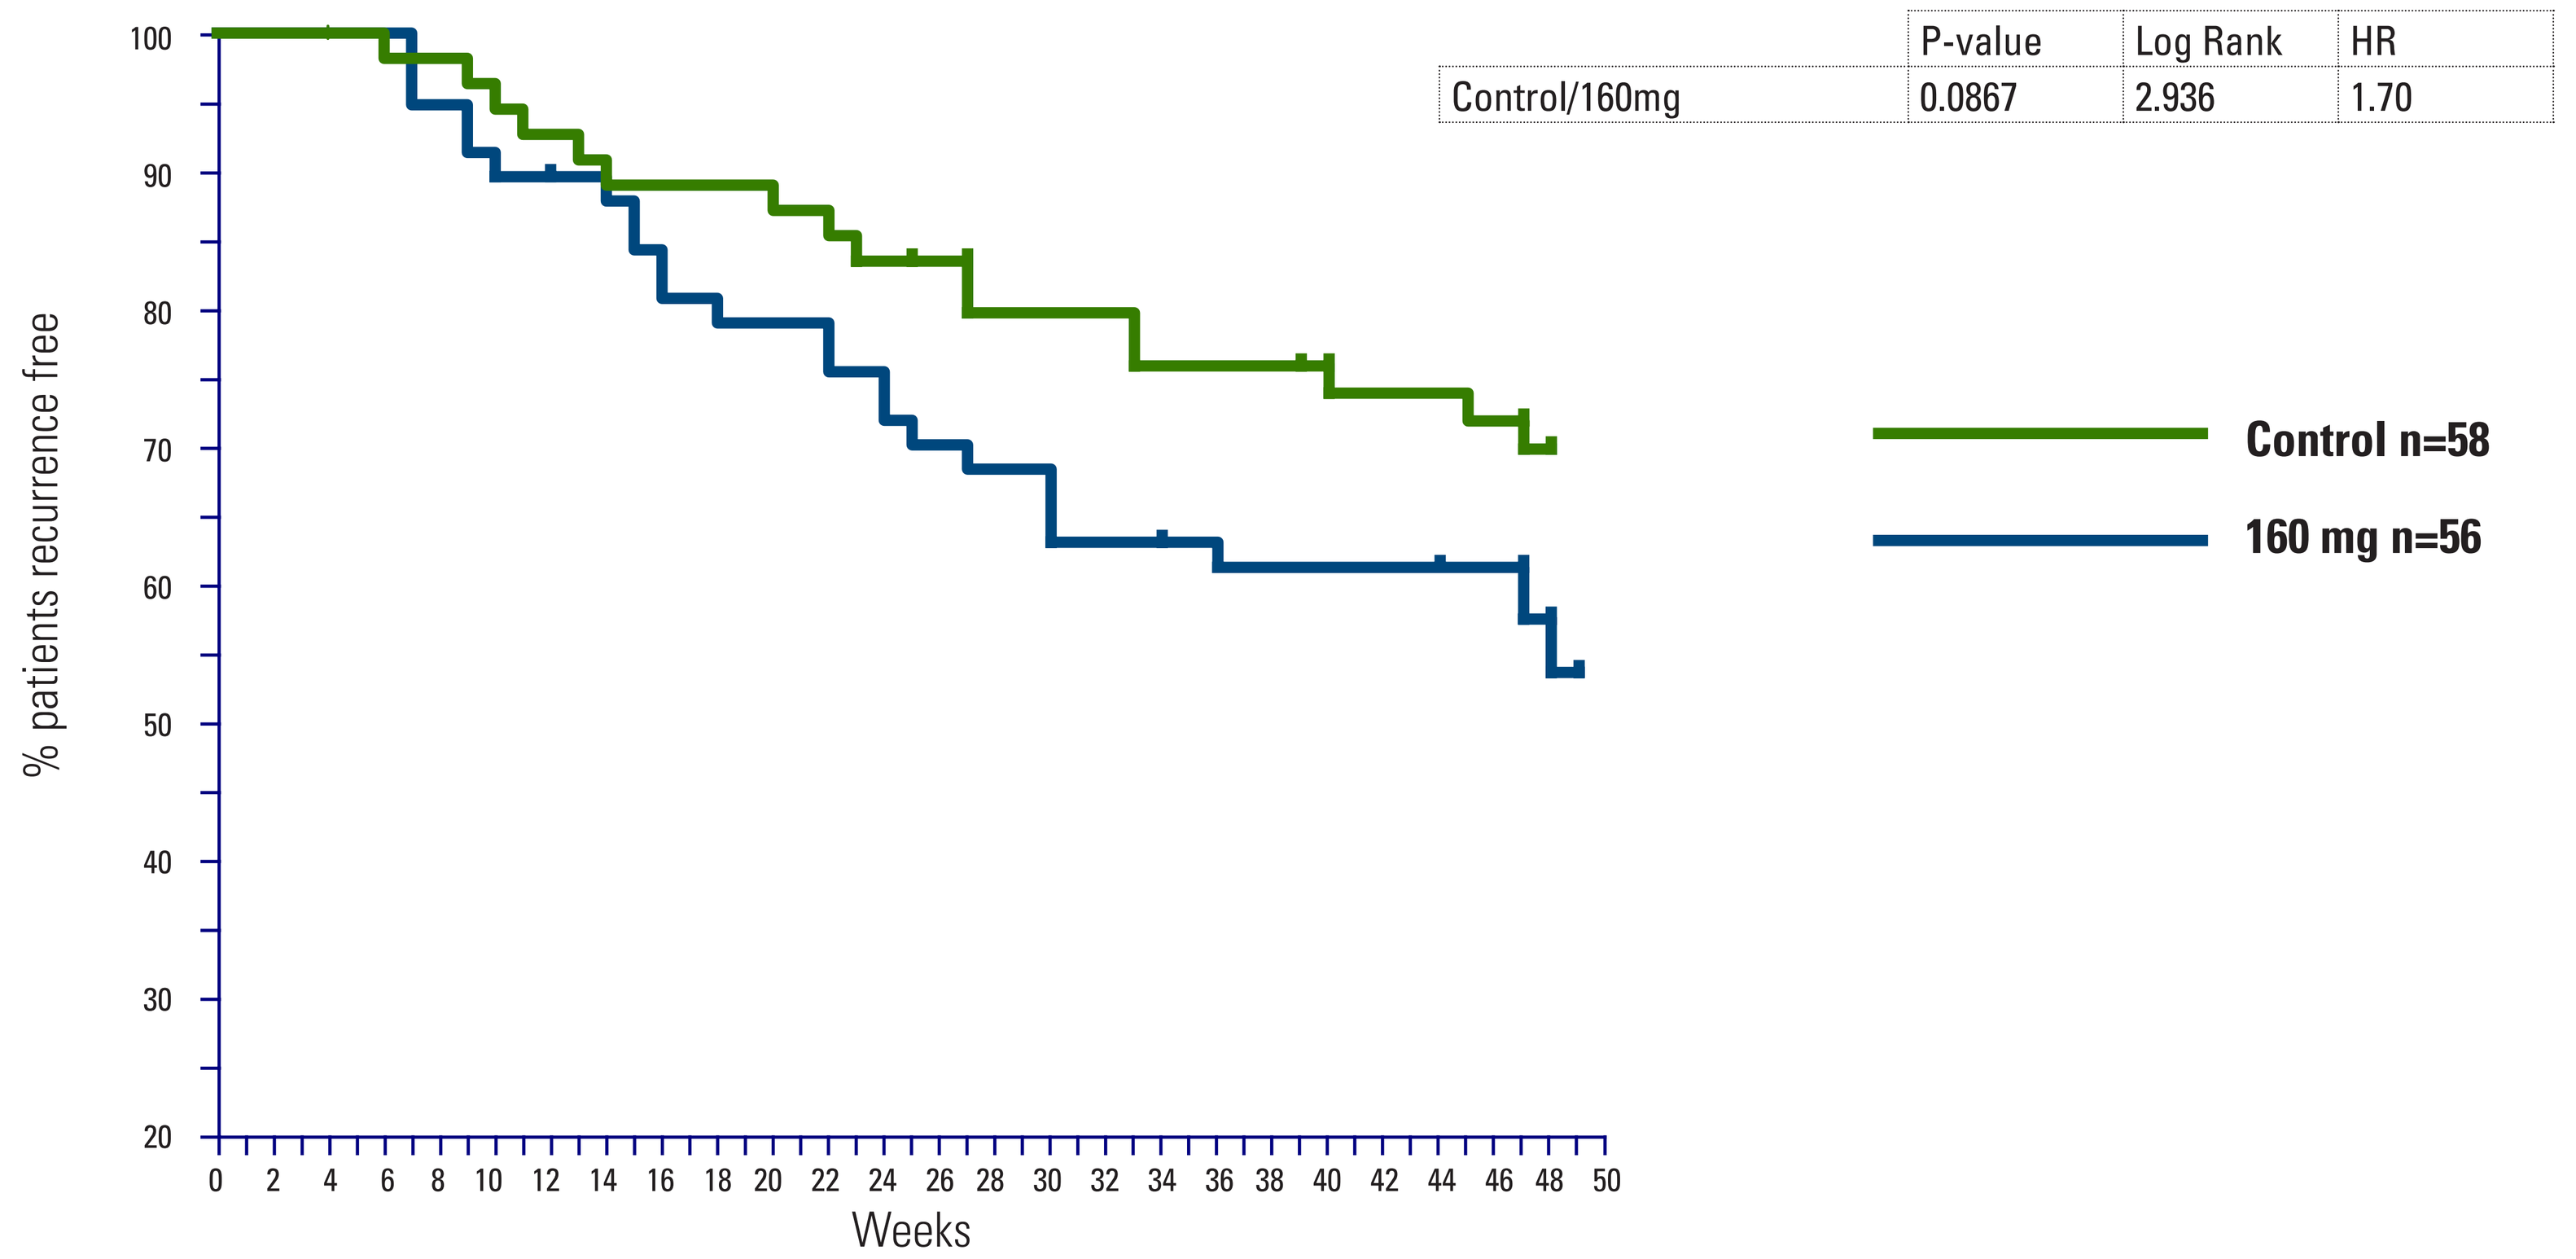
% patients recurrence free
Weeks
Control n=58
160 mg n=56

	P-value	Log Rank	HR
Control/160mg	0.0867	2.936	1.70

IMPACT OF TUMOR SIZE ON RECURRENCE RISK

The recursive partitioning analysis model - based on control arm only

Figure 5

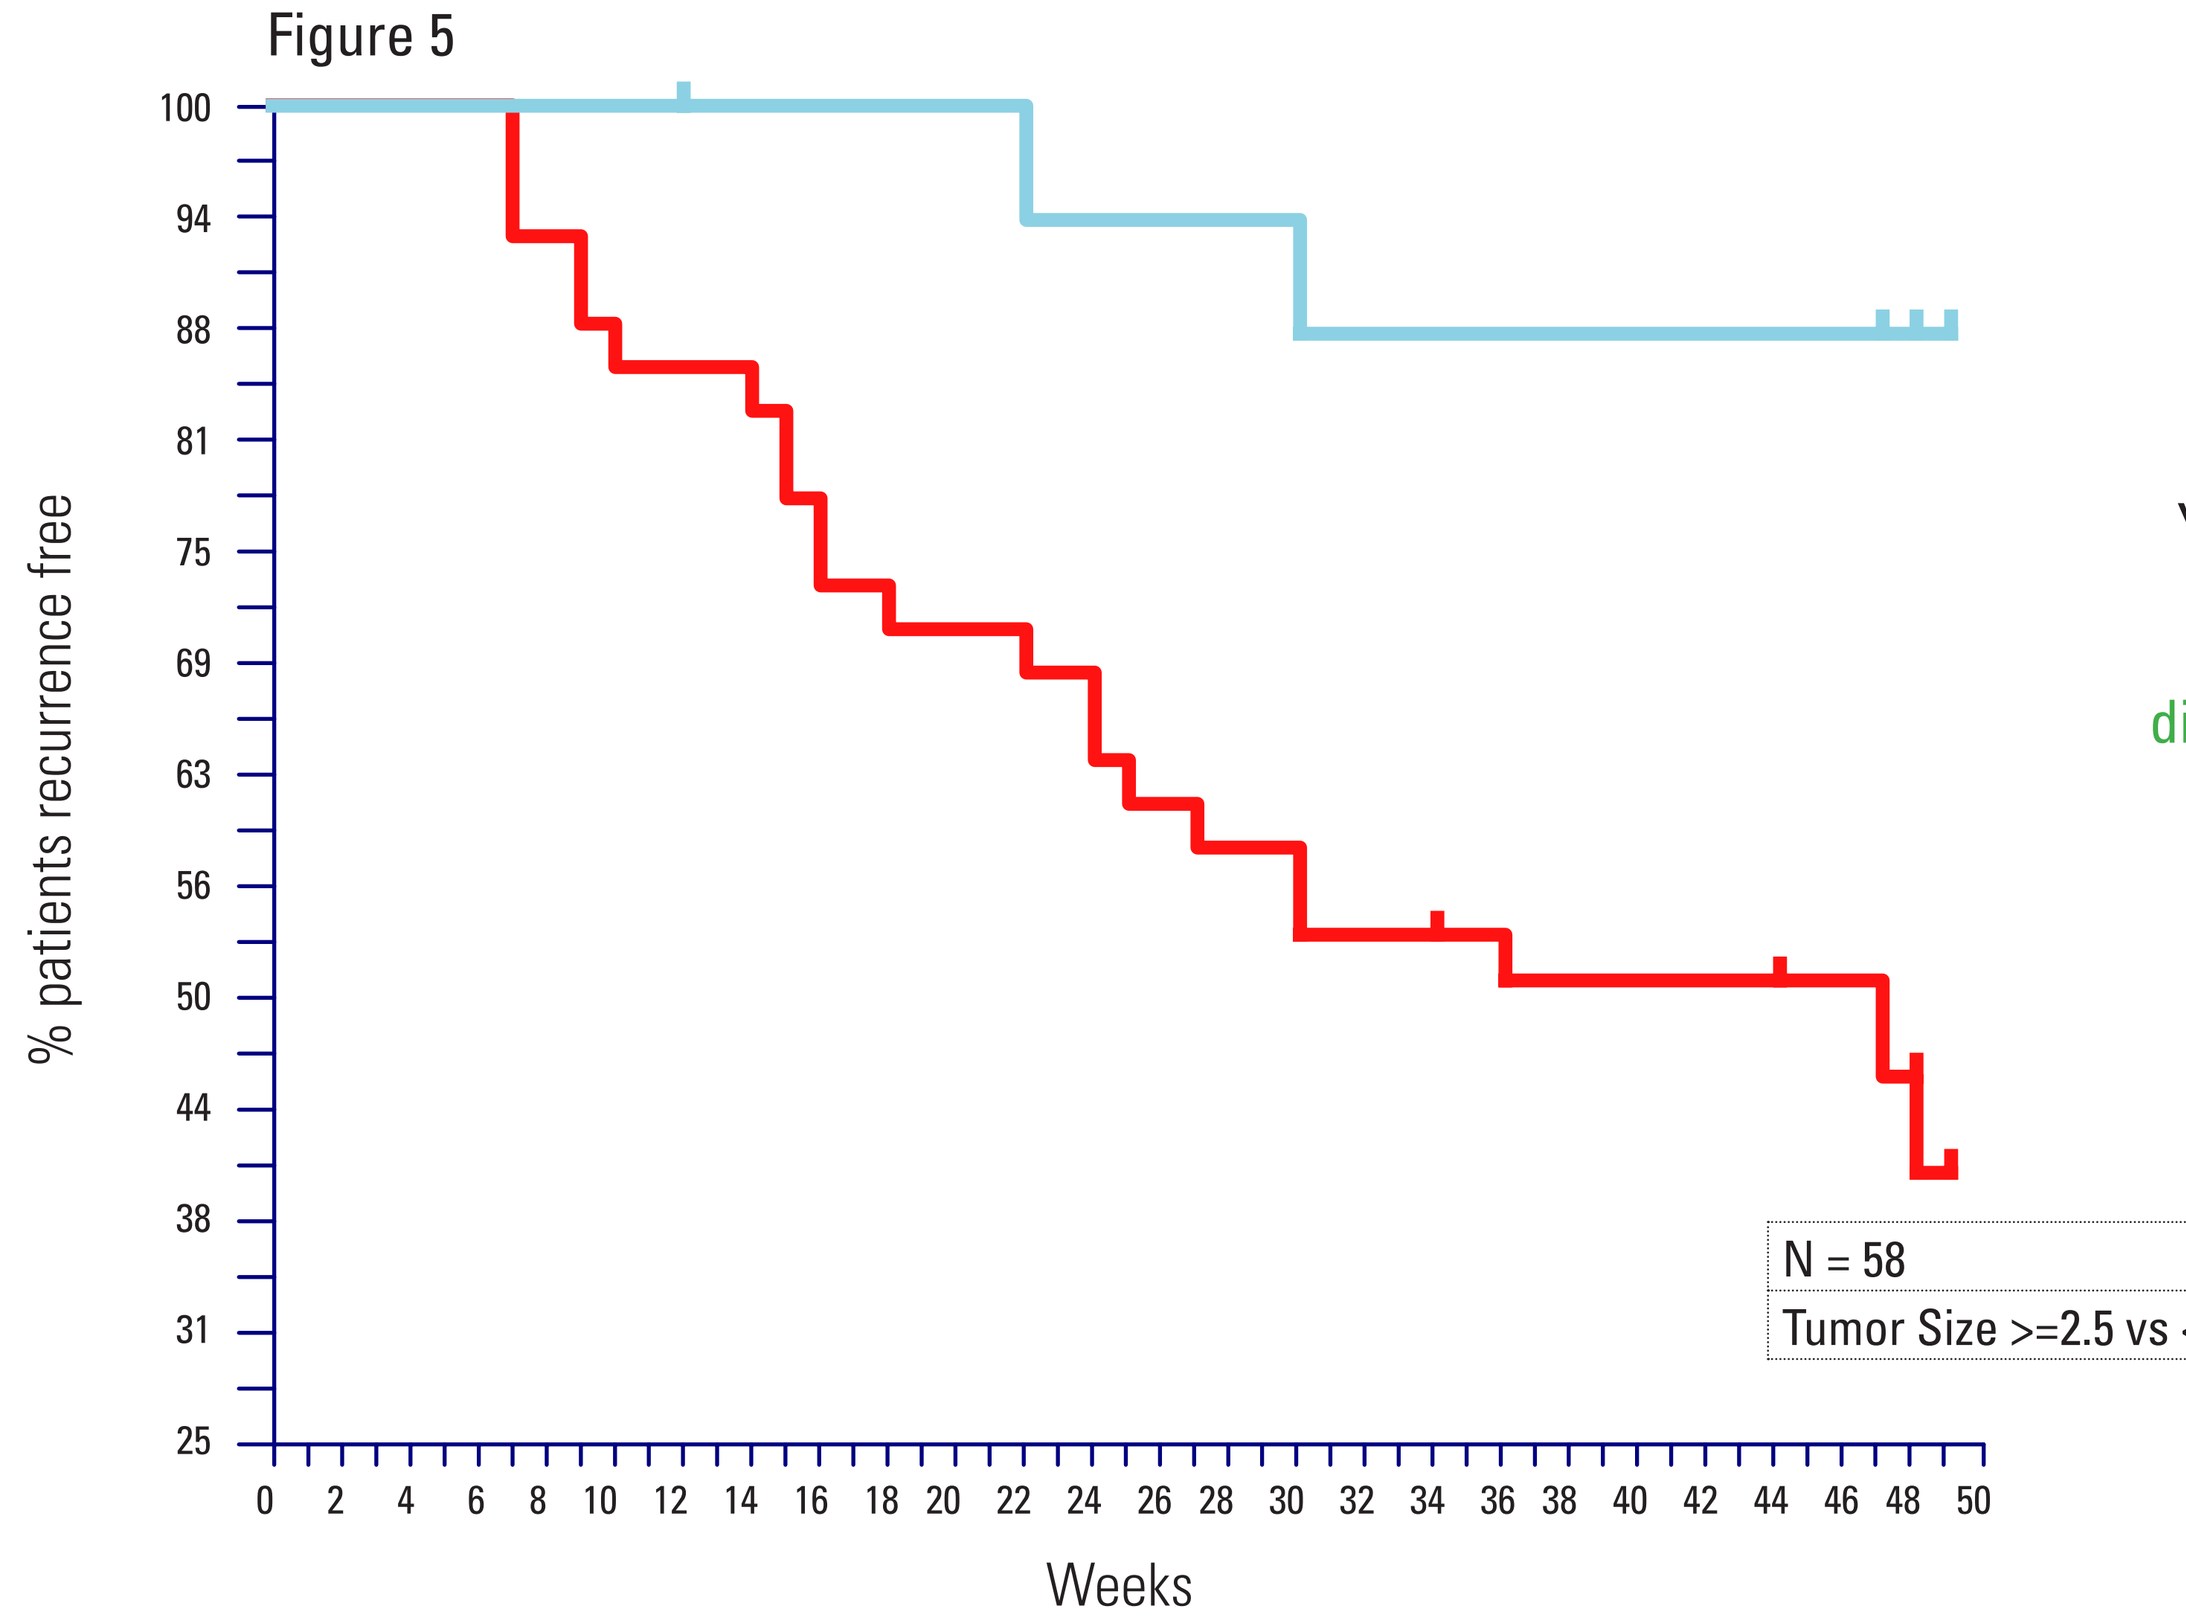
% patients recurrence free
Weeks
Tumor_size_C
>= 2.5
> 2.5

N = 58	P-value	Log Rank	HR
Tumor Size >=2.5 vs <2.5	0.0027	9.016	6.58

Tumor size is the strongest predictor in this patient population

Is tumor size < 2.5cm ?

Yes — Predict disease free

No — Other factors help predict outcome

RECURSIVE PARTITIONING ANALYSIS MODEL

The recursive partitioning analysis model - based on control arm only

Figure 6

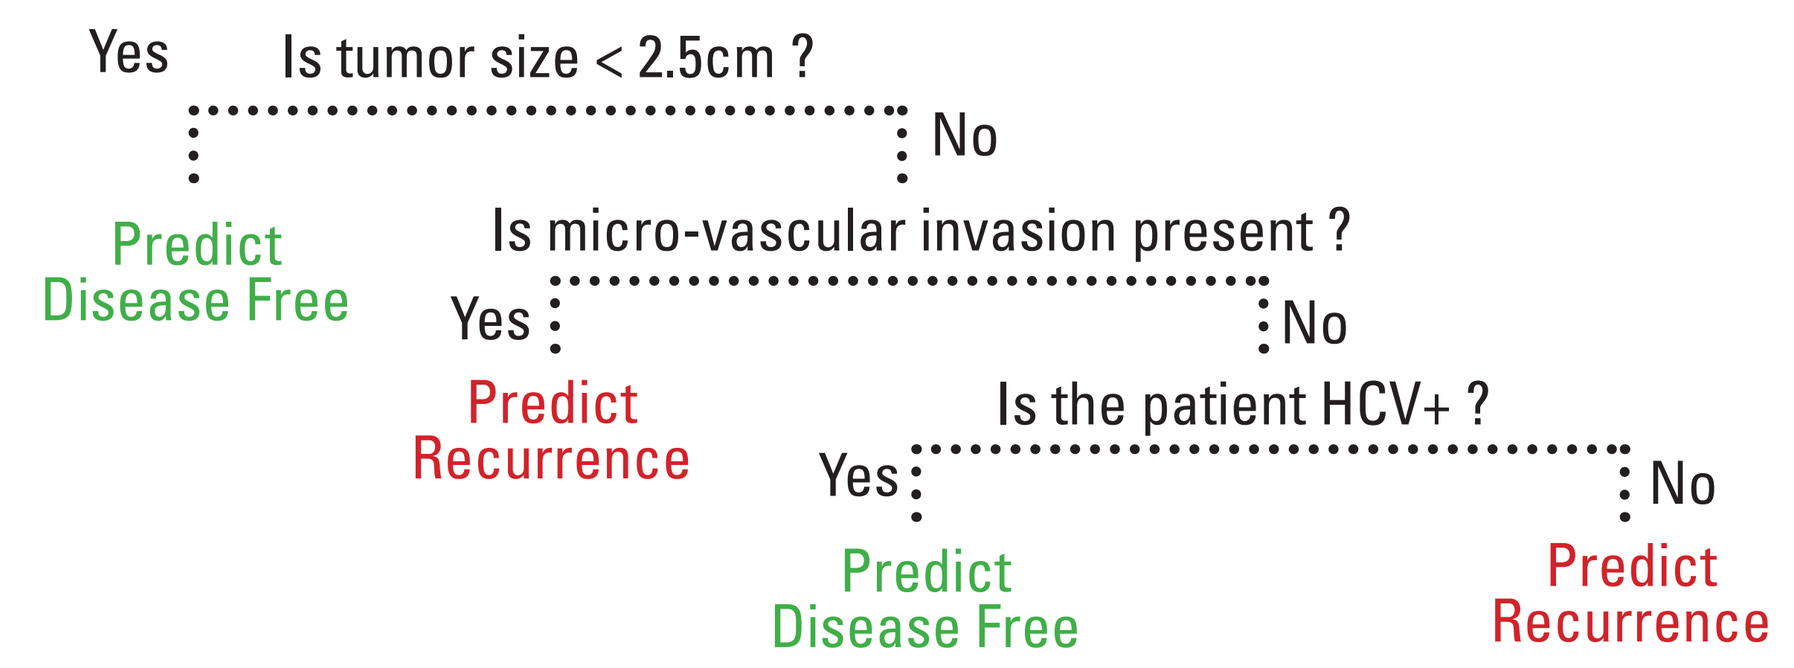
Yes
Is tumor size < 2.5cm ?
No
Predict Disease Free
Is micro-vascular invasion present ?
Yes
No
Predict Recurrence
Is the patient HCV+ ?
Yes
No
Predict Disease Free
Predict Recurrence

The number of tumors and tumor characteristics were not found to be strongly linked, in this dataset.

PI-88 TREATMENT EFFECT IN 'HIGH-RISK' SUBGROUP

Figure 7

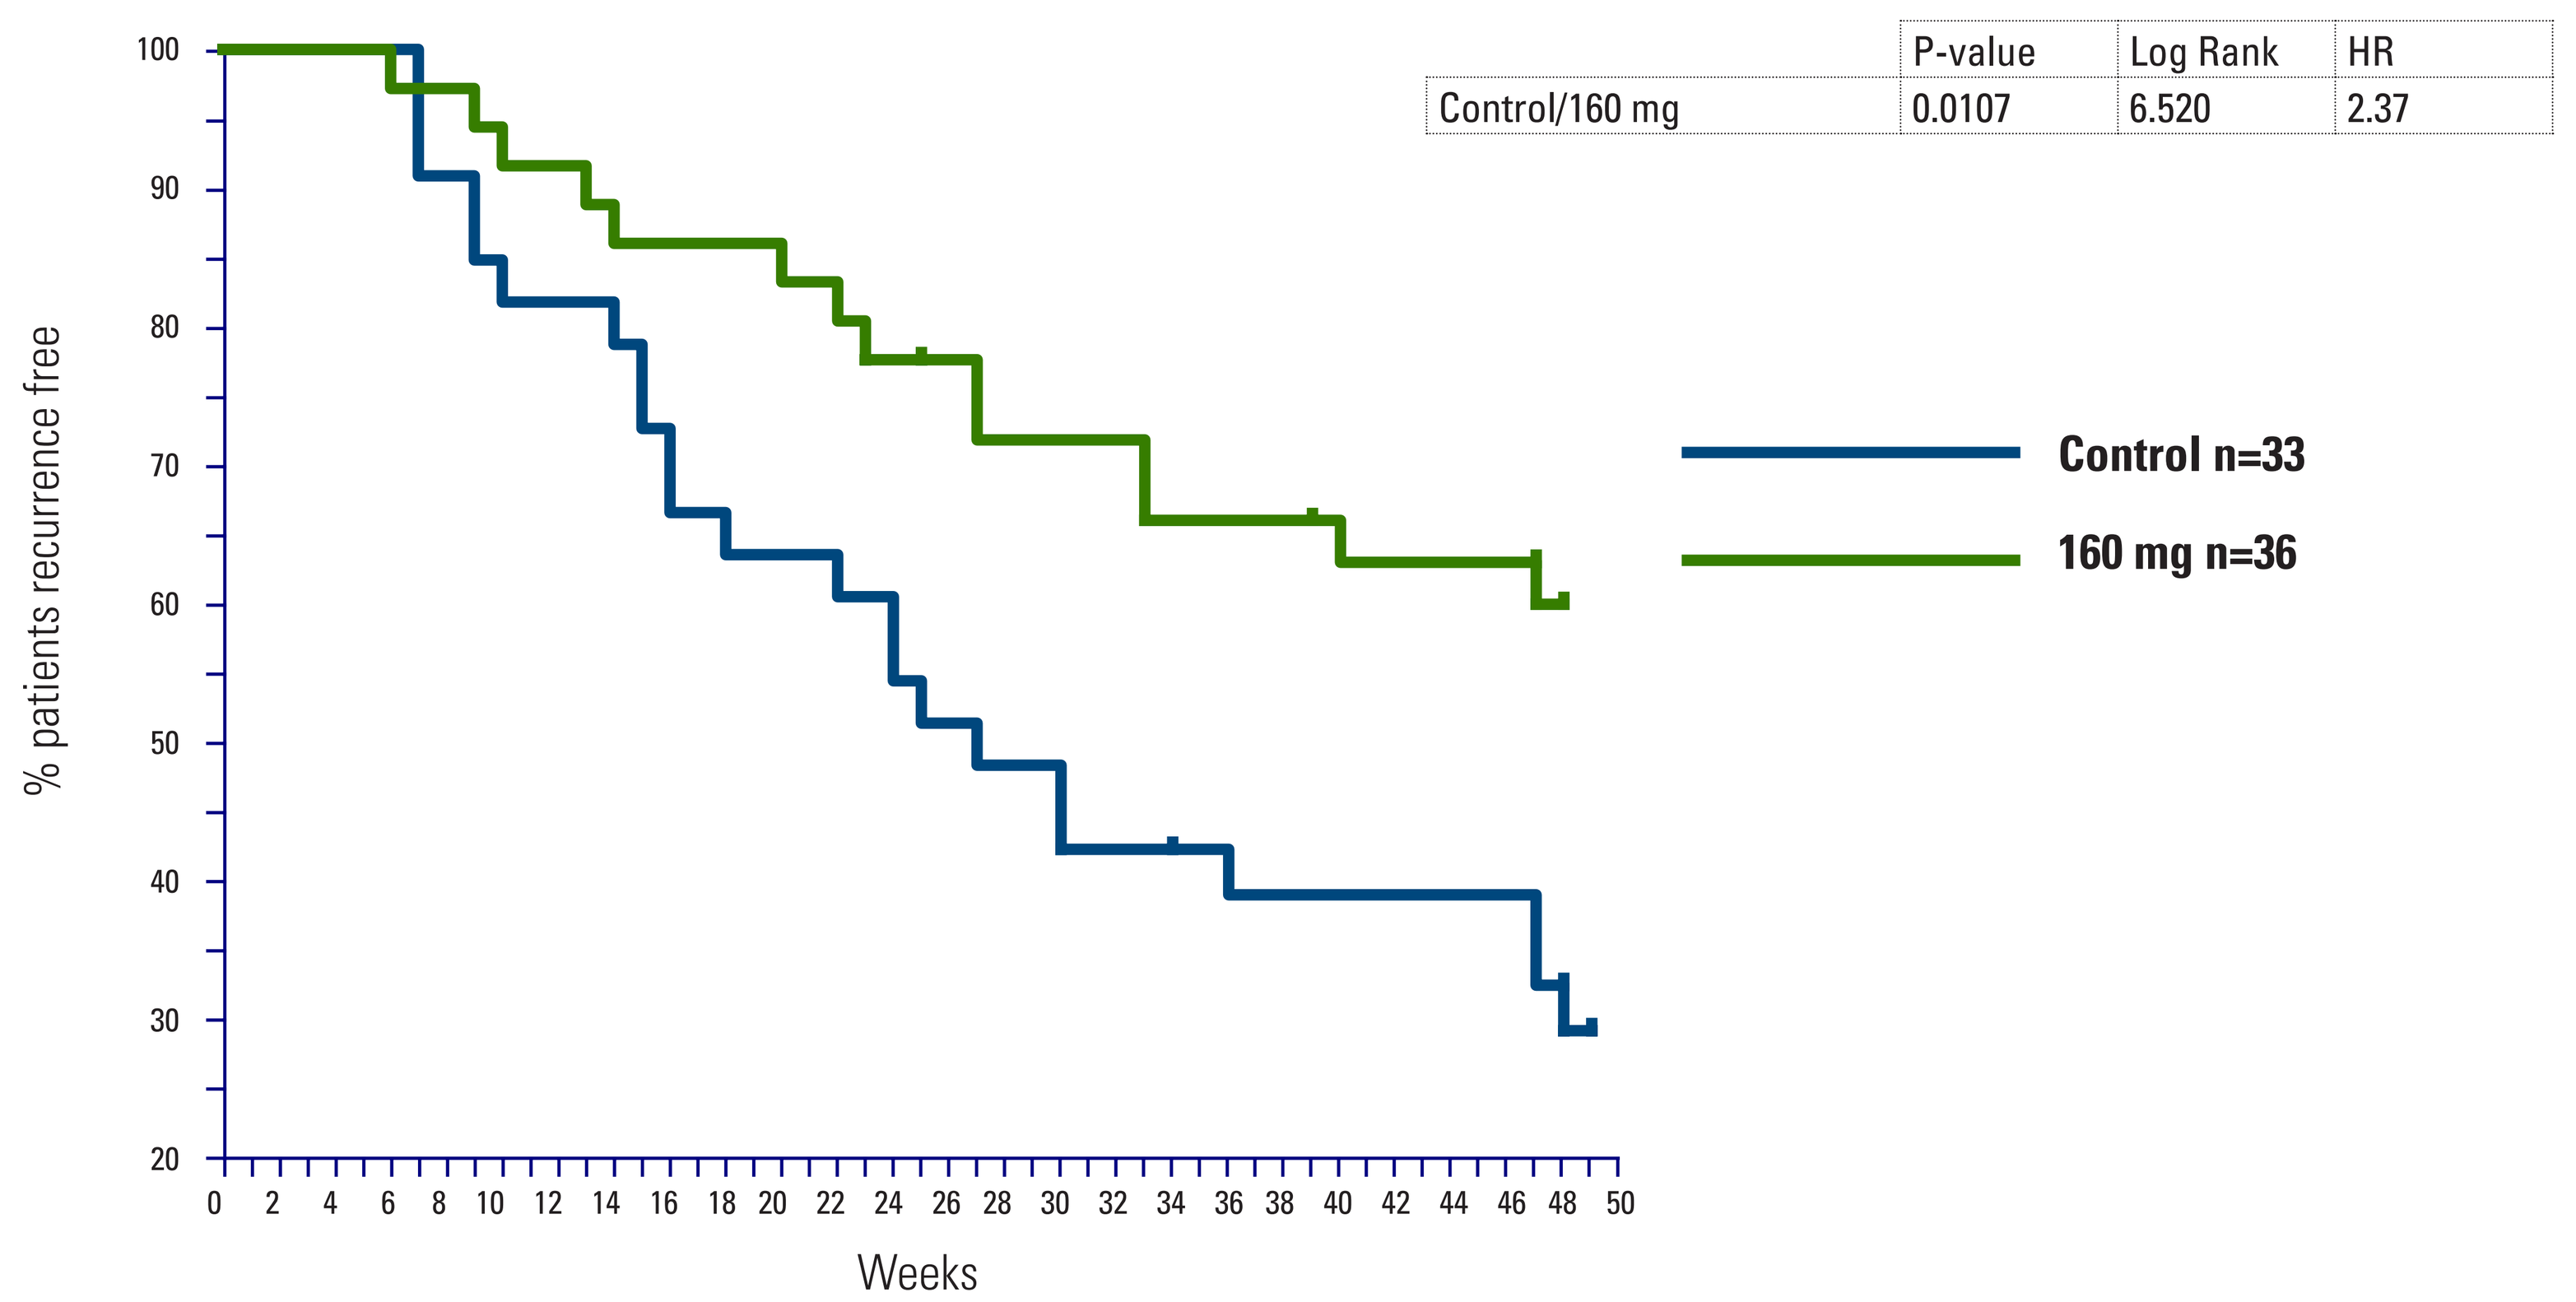
% patients recurrence free
Weeks
Control n=33
160 mg n=36

	P-value	Log Rank	HR
Control/160 mg	0.0107	6.520	2.37

The treatment effect in the' high-risk' population is similar between 160 mg and 250 mg cohorts, suggesting that apparent differences between these groups may be partly explained by differences in recurrence risk at baseline. However, the 250 mg cohort shows no evidence of incremental efficacy

CONCLUSIONS

- This analysis successfully identified a 'high-risk' population which is generally consistent with the literature
- These data better demonstrates the treatment effect of PI-88 as patients not expected to recur are excluded
- These data further support the investigation of PI-88 in an international randomized Phase 3 trial (the PATHWAY trial) assessing disease-free survival throught an event-based design

DISCLOSURE INFORMATION

The authors are employees of Progen Pharmaceuticals, the sponsor of this trial